Upregulation of Semicarbazide-Sensitive Amine Oxidase (SSAO) in Endothelial Cells Is the Mechanism of Oxidative Vascular Injury in Diabetes: Blockade by UD-014, a Novel SSAO Selective Inhibitor

Tetsuo Kawaguchi, Shinpei Nonouchi, Masaru Shinohara, Koji Itoh, and Shigeru Ushiyama, Pharmaceuticals Research Laboratory, Ube Industries, Ltd., Ube

UBE INDUSTRIES,LTD.
120th Anniversary

Introduction

Semicarbazide-sensitive amine oxidase (SSAO) / vascular adhesion protein-1 (VAP-1) expressed on the endothelium catalyzes deamination of primary monoamines[1].

SSAO

$CH_3NH_2 + O_2 + H_2O \Rightarrow HCHO + H_2O_2 + NH_3$

$CH_3CO\ CH_2NH_2 + O_2 + H_2O \Rightarrow CH_3COCHO + H_2O_2 + NH_3$

This enzyme also mediates leukocyte rolling, adhesion and transmigration through the endothelium[2,3].

SSAO activity is shed from the endothelium into plasma and is known to be elevated in diabetes mellitus[4,5]. Thus, SSAO has been considered to be one of the causative factors to induce microvascular injury in diabetes[6]. Our recent study found that UD-014, a novel SSAO selective inhibitor, ameliorated albuminuria in the streptozotocin-induced diabetic nephropathy model in rats (Refer to 503-P at ADA2017). However, its mechanism remains unclear. The aim of this study is to clarify the mechanism of action of UD-014 using glomerular microvascular endothelial cells (GMVECs) focusing on the vascular injury induced by oxidative stress.

UD-014 *in vitro* profile

IC_{50} [nM]	Human recombinant			
	SSAO	MAO-A	MAO-B	DAO
UD-014	3.2	6,100	12,000	690
Mofegiline	5.2	550	0.51	ND
Clorgyline	ca.10,000	2.6	4,100	ND
Pargyline	ca.10,000	2,200	73	ND

Materials and Methods

- Human GMVECs (DS Pharma BioMedical) were cultured in exclusive culture medium with 10% FBS in type-I collagen coated culture ware. Cells were subjected to one day preculture before the experiments.
- Methylamine-induced cytotoxicity assay was performed[7]. Cell survival was detected by a WST-8 reduction assay.
- In quantitative RT-PCR analysis, the threshold cycle (Ct) was used for determining the relative expression level of each gene, by normalizing to the Ct of GAPDH.
- In Immunocytochemistry, cells were fixed by 4% PFA, permeabilized with 0.1% Triton X-100, immunostained with anti-human SSAO antibody (Santa Cruz) and Alexa647-conjugated secondary antibody, and imaged with a fluorescent microscope.

Methylamine induced Cytotoxicity

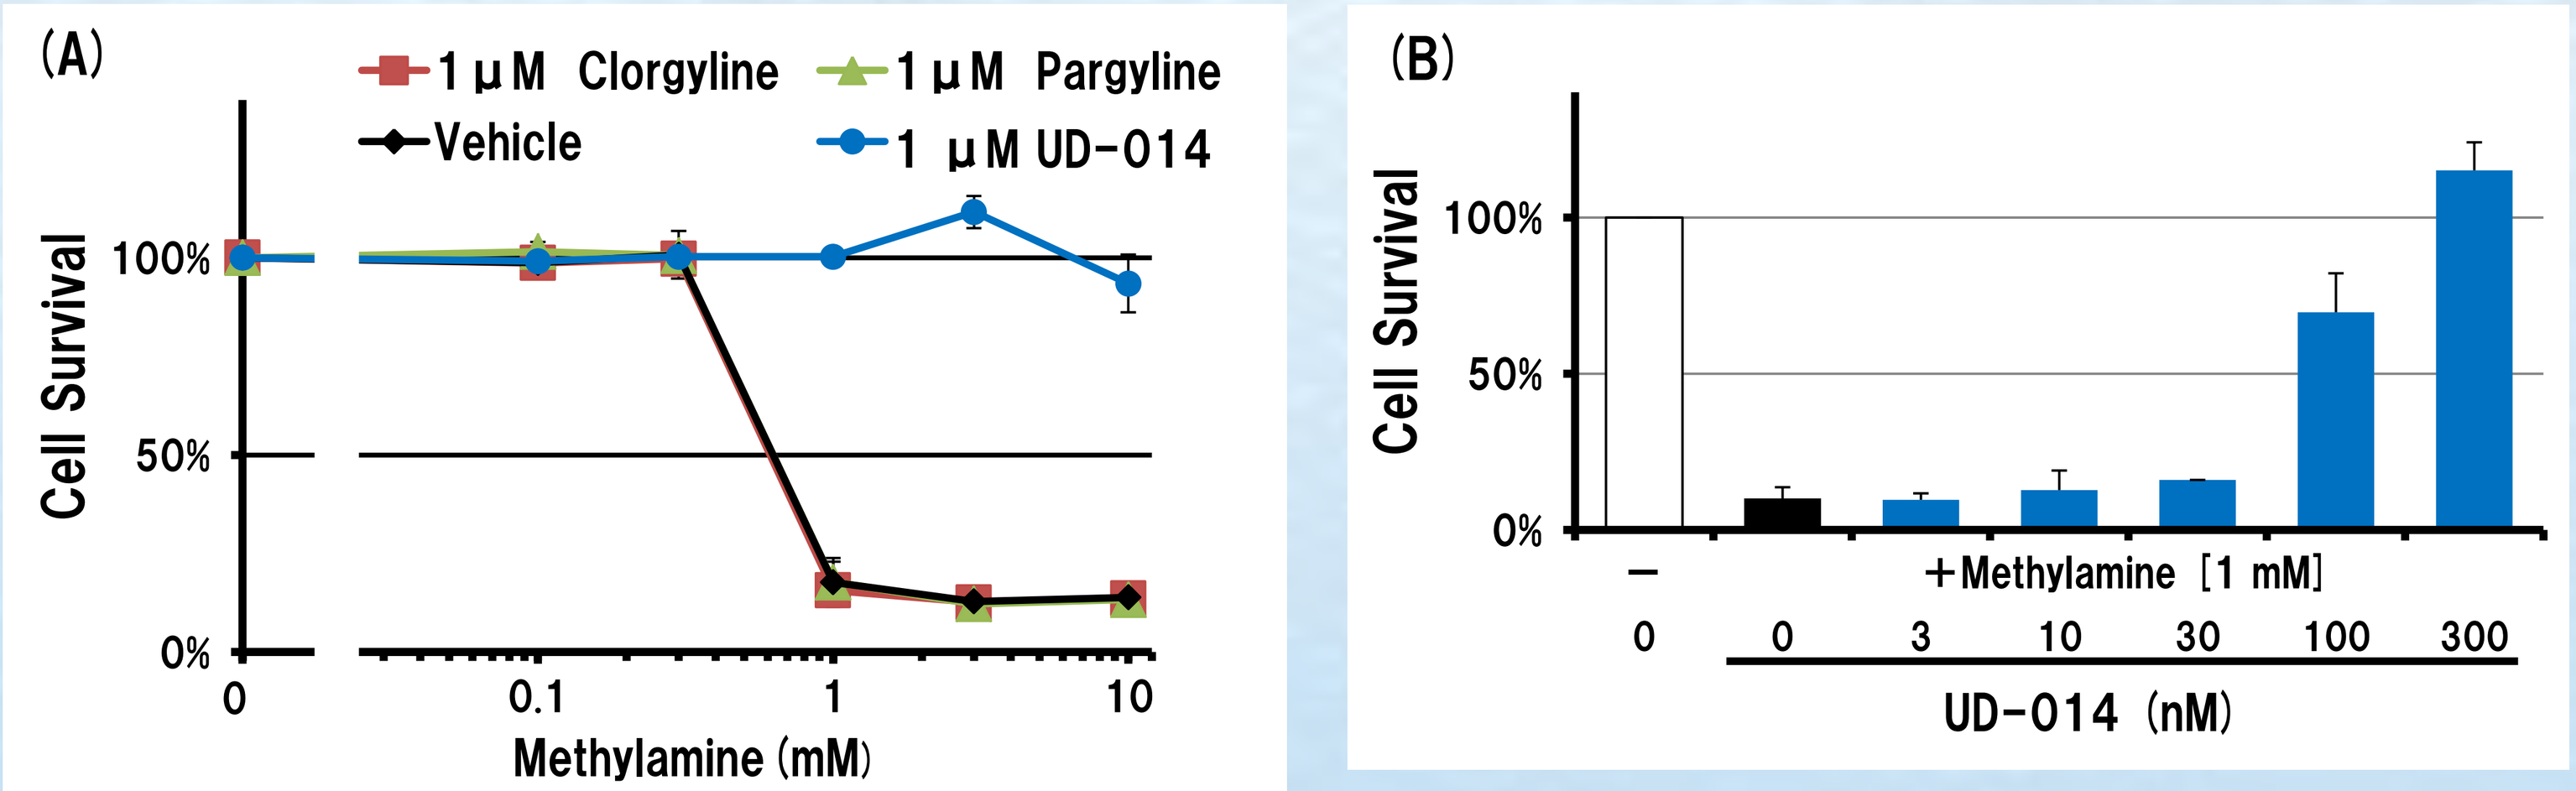
(A)
1 μM Clorgyline
1 μM Pargyline
Vehicle
1 μM UD-014
Cell Survival
Methylamine (mM)
(B)
Cell Survival
+Methylamine [1 mM]
UD-014 (nM)

Fig.1 Methylamine induced cytotoxicity during 2-day culture. (A) Methylamine at 1 mM and more induced cytotoxicity in GMVECs. An SSAO inhibitor, UD-014 at 1 μM completely rescued the toxicity. Neither MAO-A (Red) nor MAO-B (Green) inhibitors did. (B) UD-014 rescued the methylamine-induced cytotoxicity concentration-dependently.

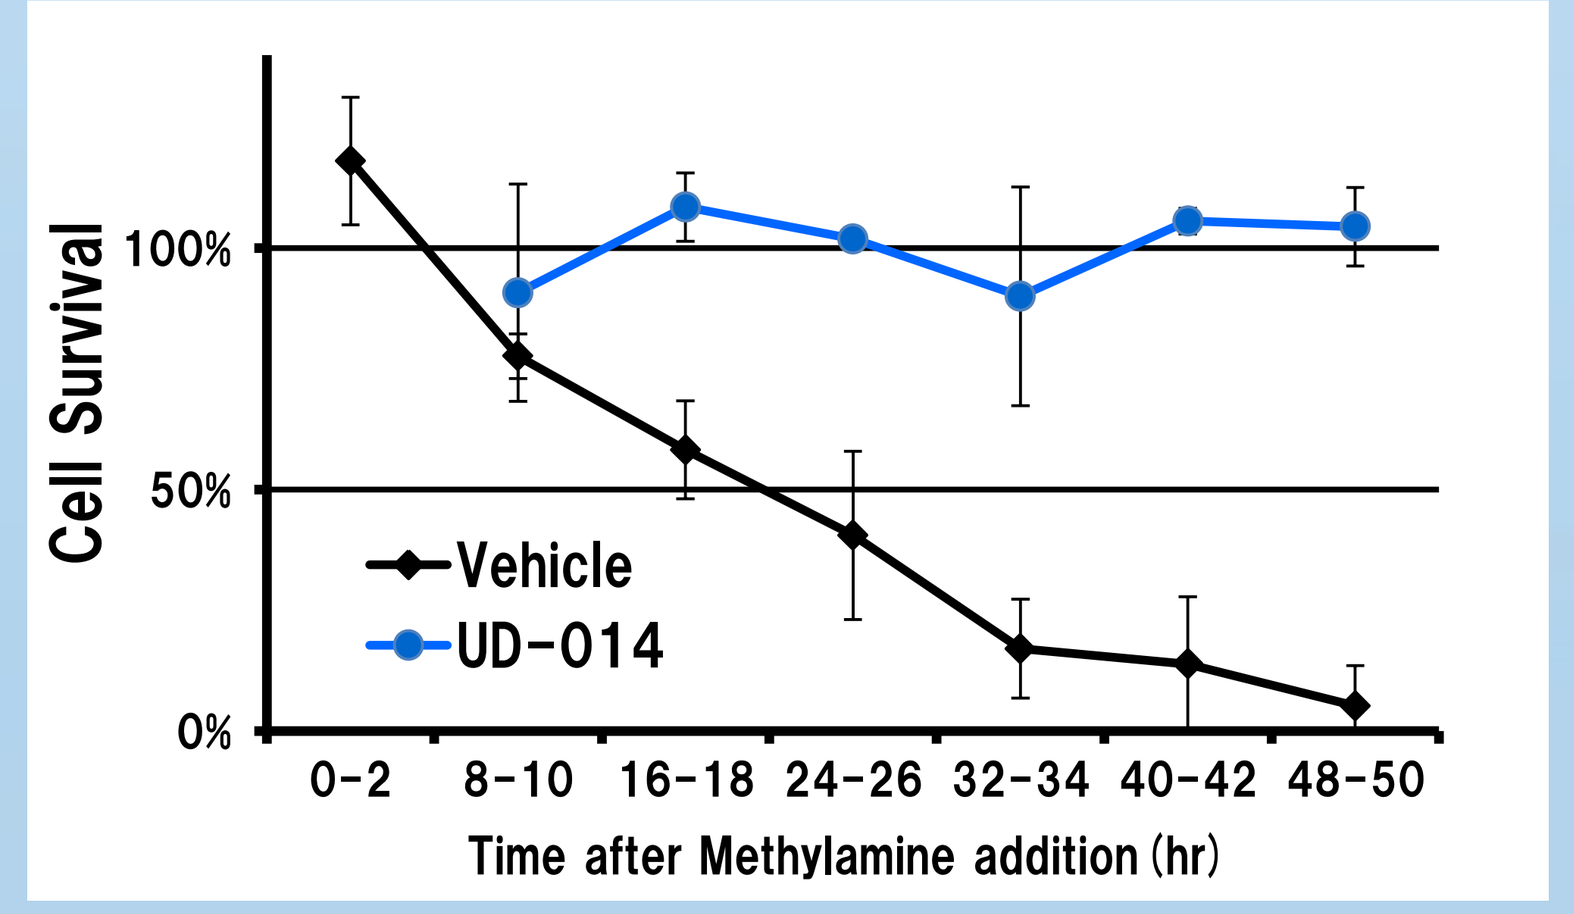
Cell Survival
Vehicle
UD-014
Time after Methylamine addition (hr)

Fig.2 Time dependent cytotoxicity by 1 mM methylamine. The cell death progressed slowly, and 1 μM UD-014 inhibited it completely.

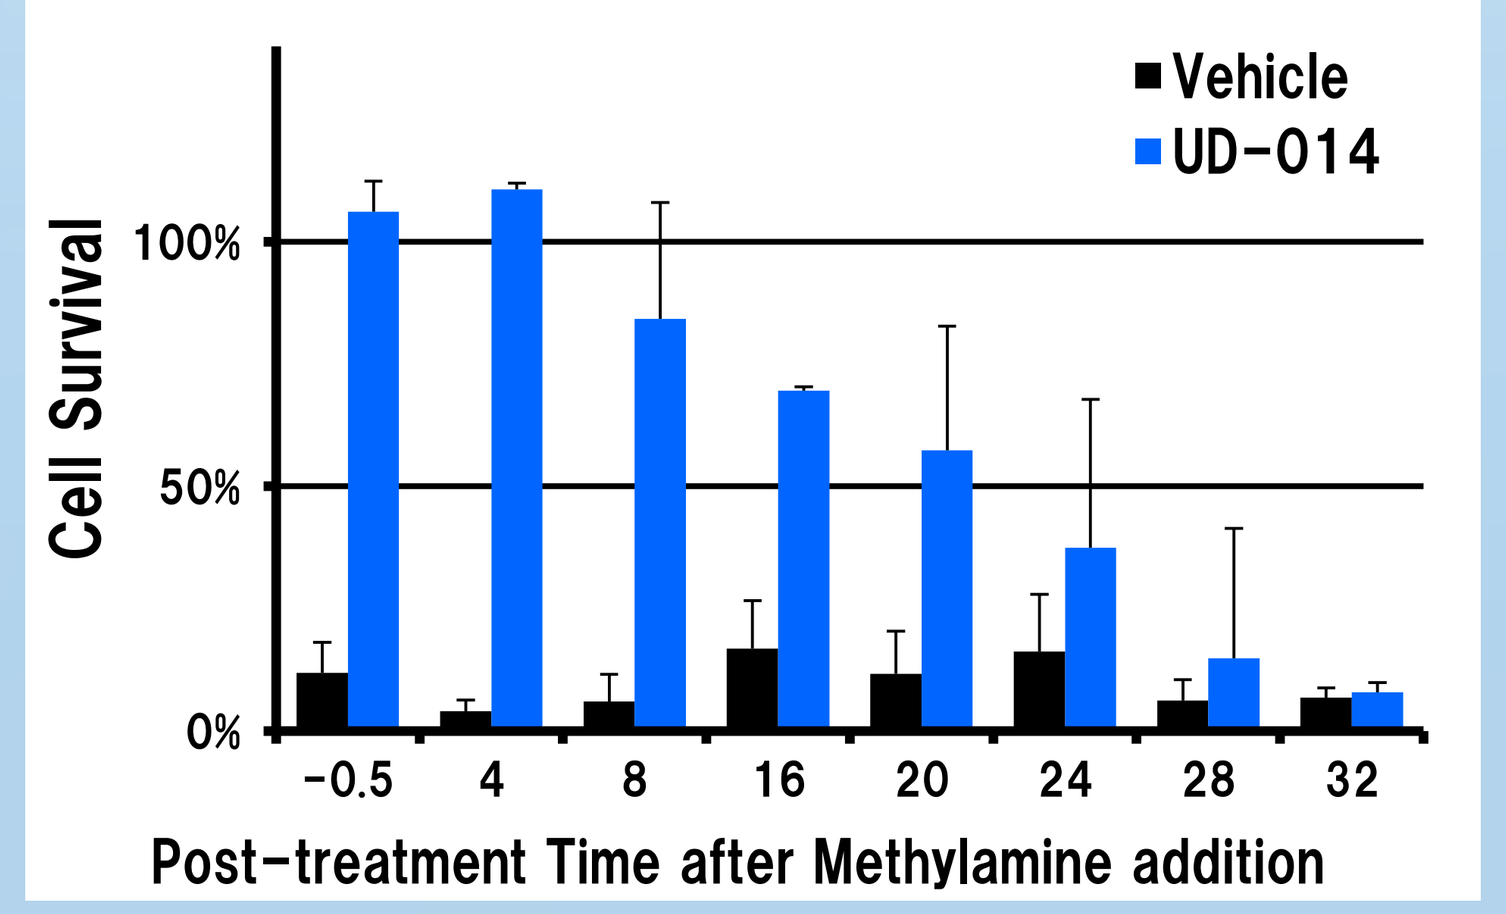
Vehicle
UD-014
Cell Survival
Post-treatment Time after Methylamine addition

Fig.3 Post-treatment with UD-014. Until 20 hrs post-treatment, 1 μM UD-014 rescued more than 50% of cells in 2-day culture.

SSAO mRNA Induction

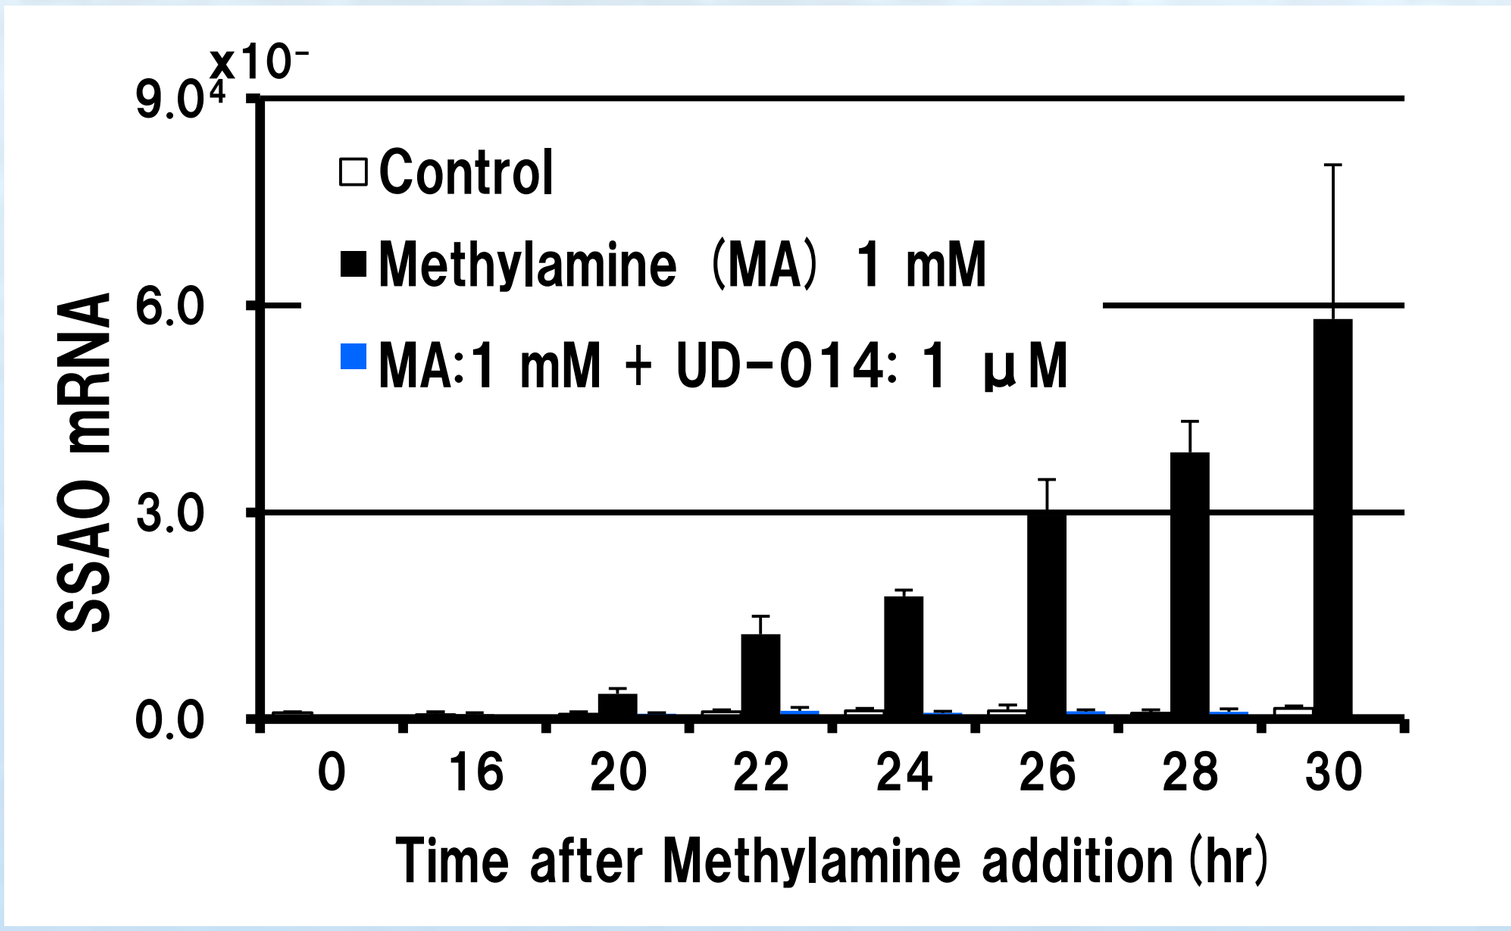
Control
Methylamine (MA) 1 mM
MA:1 mM + UD-014: 1 μM
SSAO mRNA
Time after Methylamine addition (hr)

Fig.5 Methylamine induced SSAO mRNA upregulation. Methylamine induced the mRNA more than 50-fold after 30 hr incubation. UD-014 completely inhibited the induction.

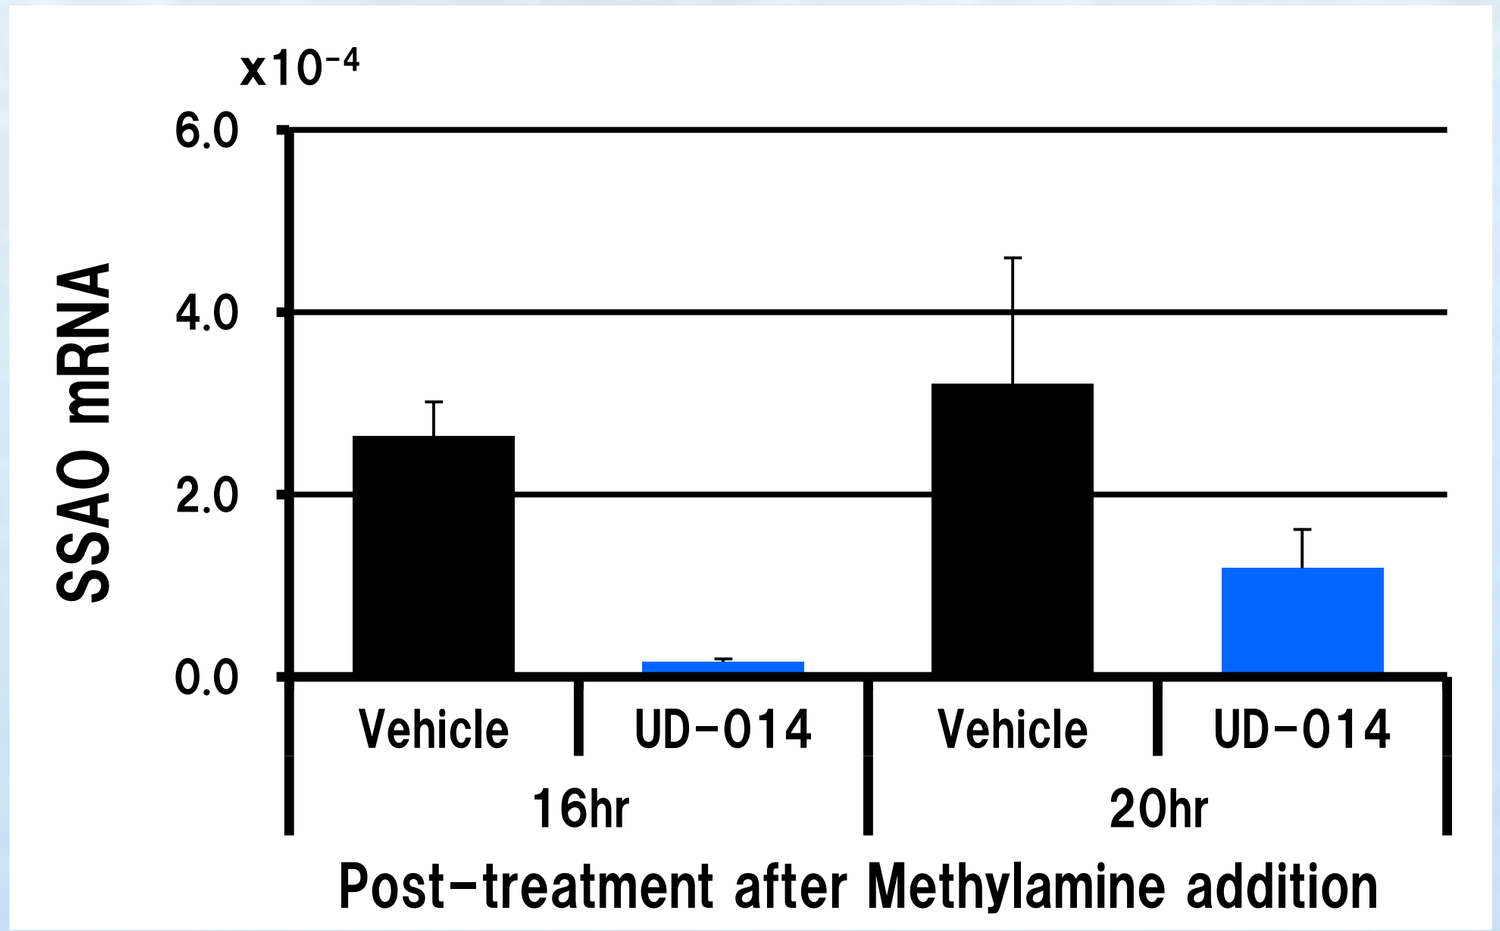
SSAO mRNA
Vehicle
UD-014
16hr
20hr
Post-treatment after Methylamine addition

Fig.6 Post-treatment with UD-014. Even after 16 hr post-treatment with 1 μM UD-014 inhibited SSAO mRNA induction.

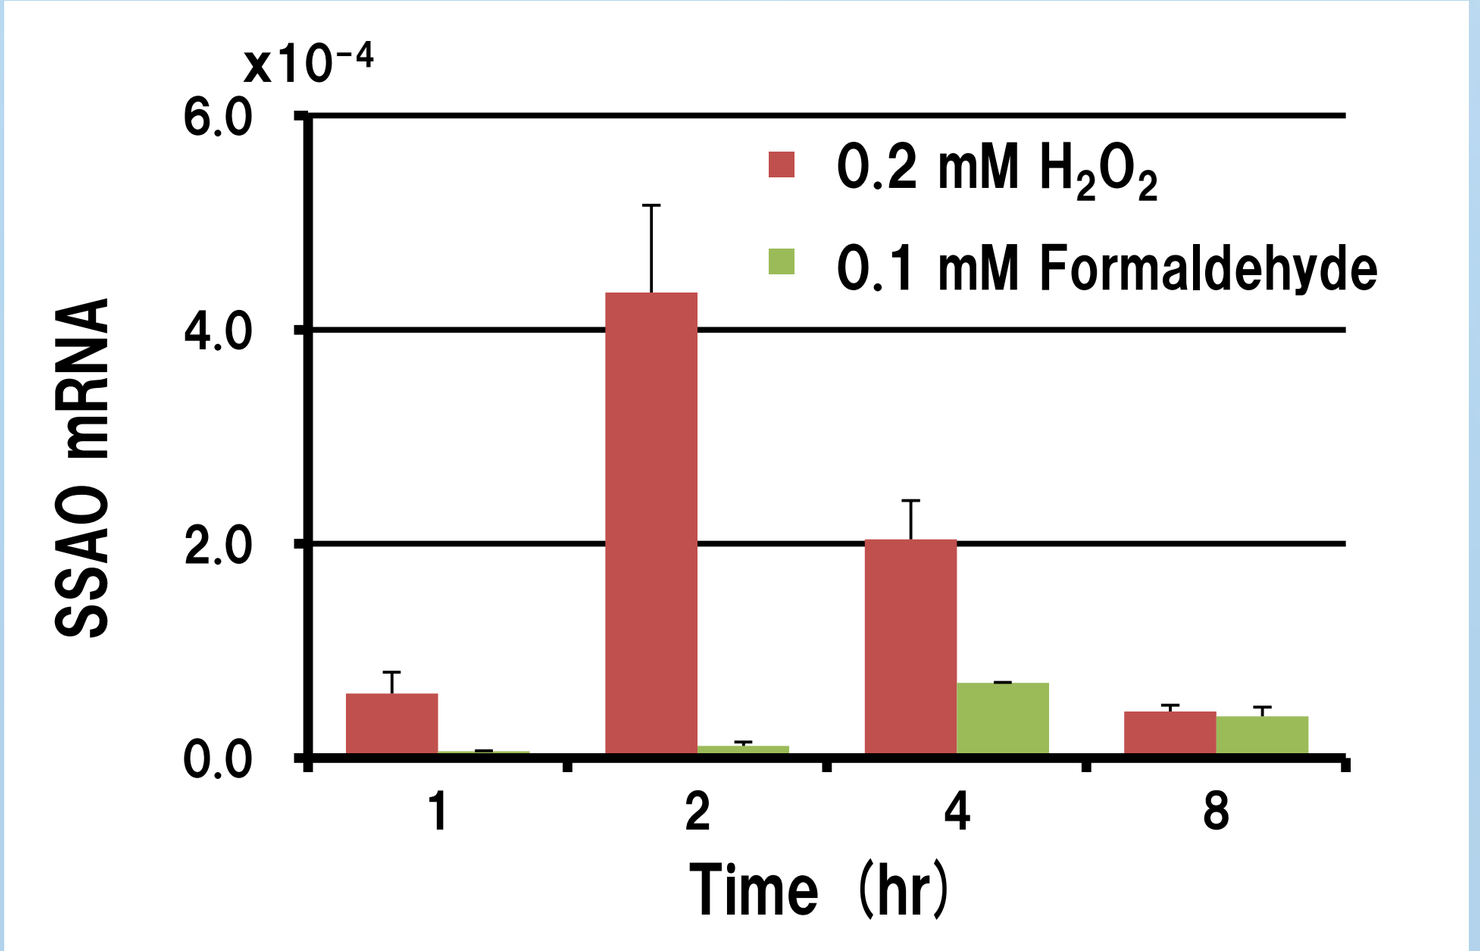
0.2 mM H2O2
0.1 mM Formaldehyde
SSAO mRNA
Time (hr)

Fig.7 H_2O_2 induced SSAO mRNA. H_2O_2 upregulated the mRNA quickly and strongly. The upregulation of formaldehyde was weak.

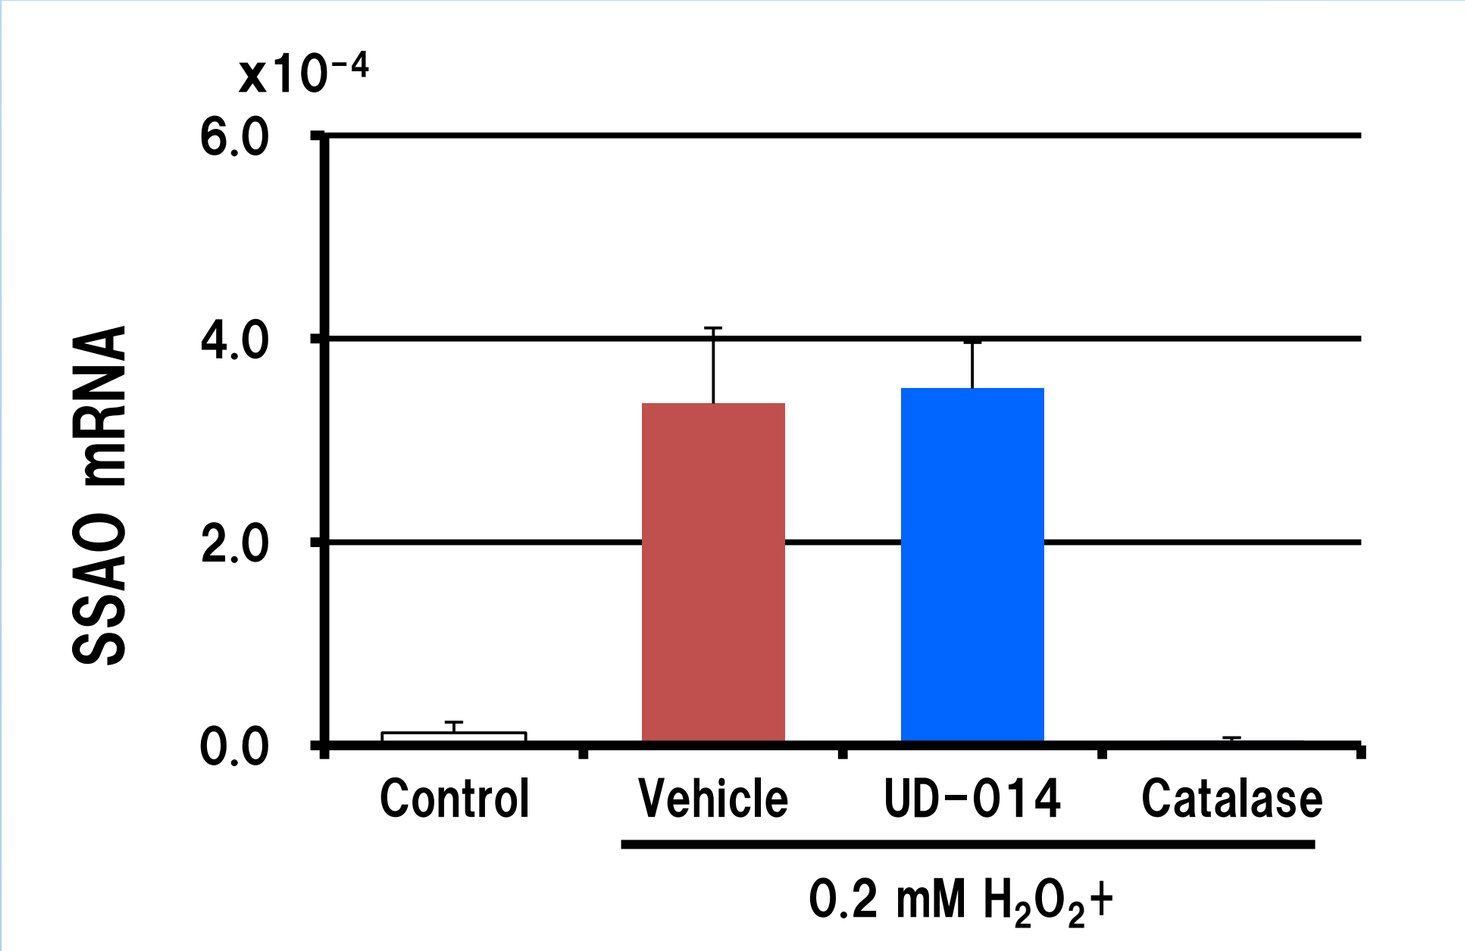
SSAO mRNA
Control
Vehicle
UD-014
Catalase
0.2 mM H2O2+

Fig.8 SSAO mRNA induction by H_2O_2 in 2 hrs. 1 μM UD-014 had no effect on H_2O_2–induced mRNA upregulation.

Gene Expression Change

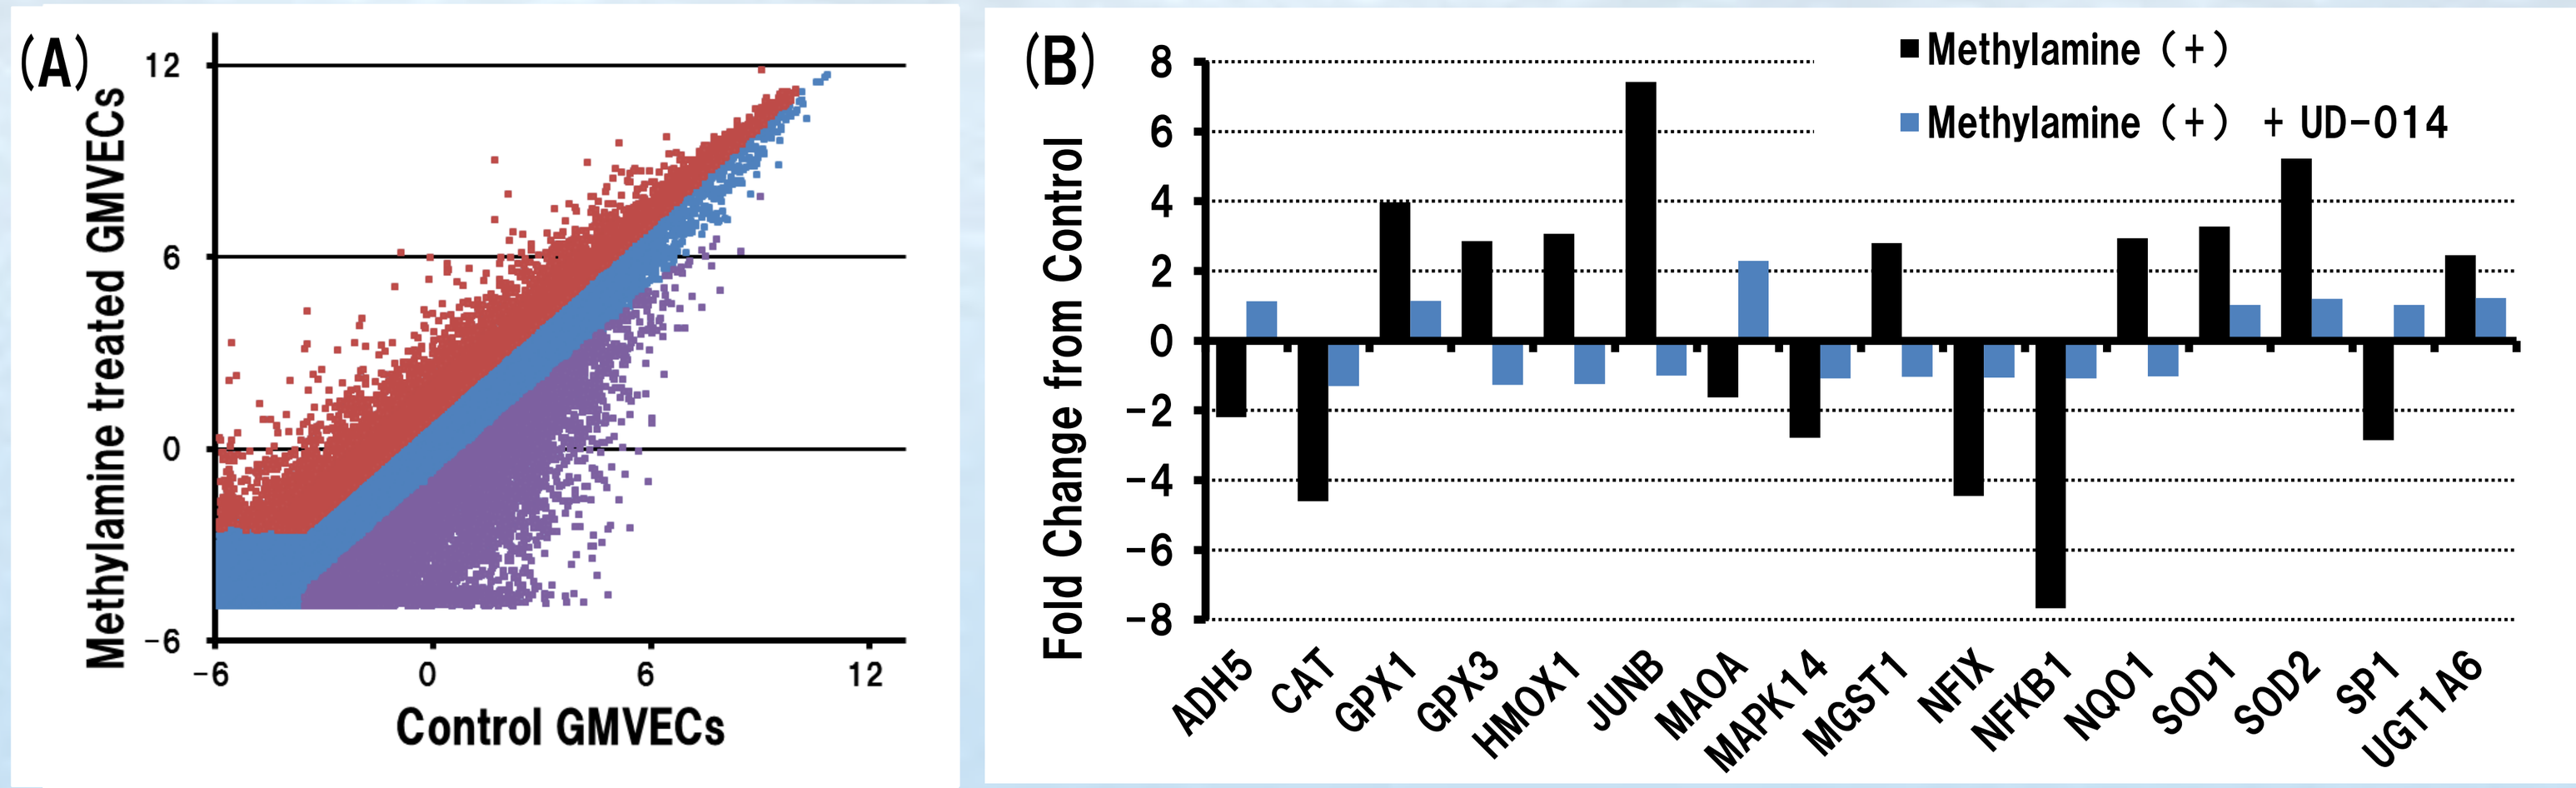
(A)
Methylamine treated GMVECs
Control GMVECs
(B)
Methylamine (+)
Methylamine (+) + UD-014
Fold Change from Control
ADH5
CAT
GPX1
GPX3
HMOX1
JUNB
MAOA
MAPK14
MGST1
NFIX
NFKB1
NQO1
SOD1
SOD2
SP1
UGT1A6

Fig.10 Microarray analysis of gene expression in methylamine treated GMVECs. (A) Scatter plot shows the 2-fold upregulated or down-regulated genes (the red and purple dots, respectively). (B) Effect of UD-014 treatment on the ADH5 (formaldehyde dehydrogenase) and oxidative stress pathway genes expression. The results were analyzed using Agilent SurePrint G3 Human V2 GE 8X60K Microarray.

Methylamine Metabolites Cytotoxicity

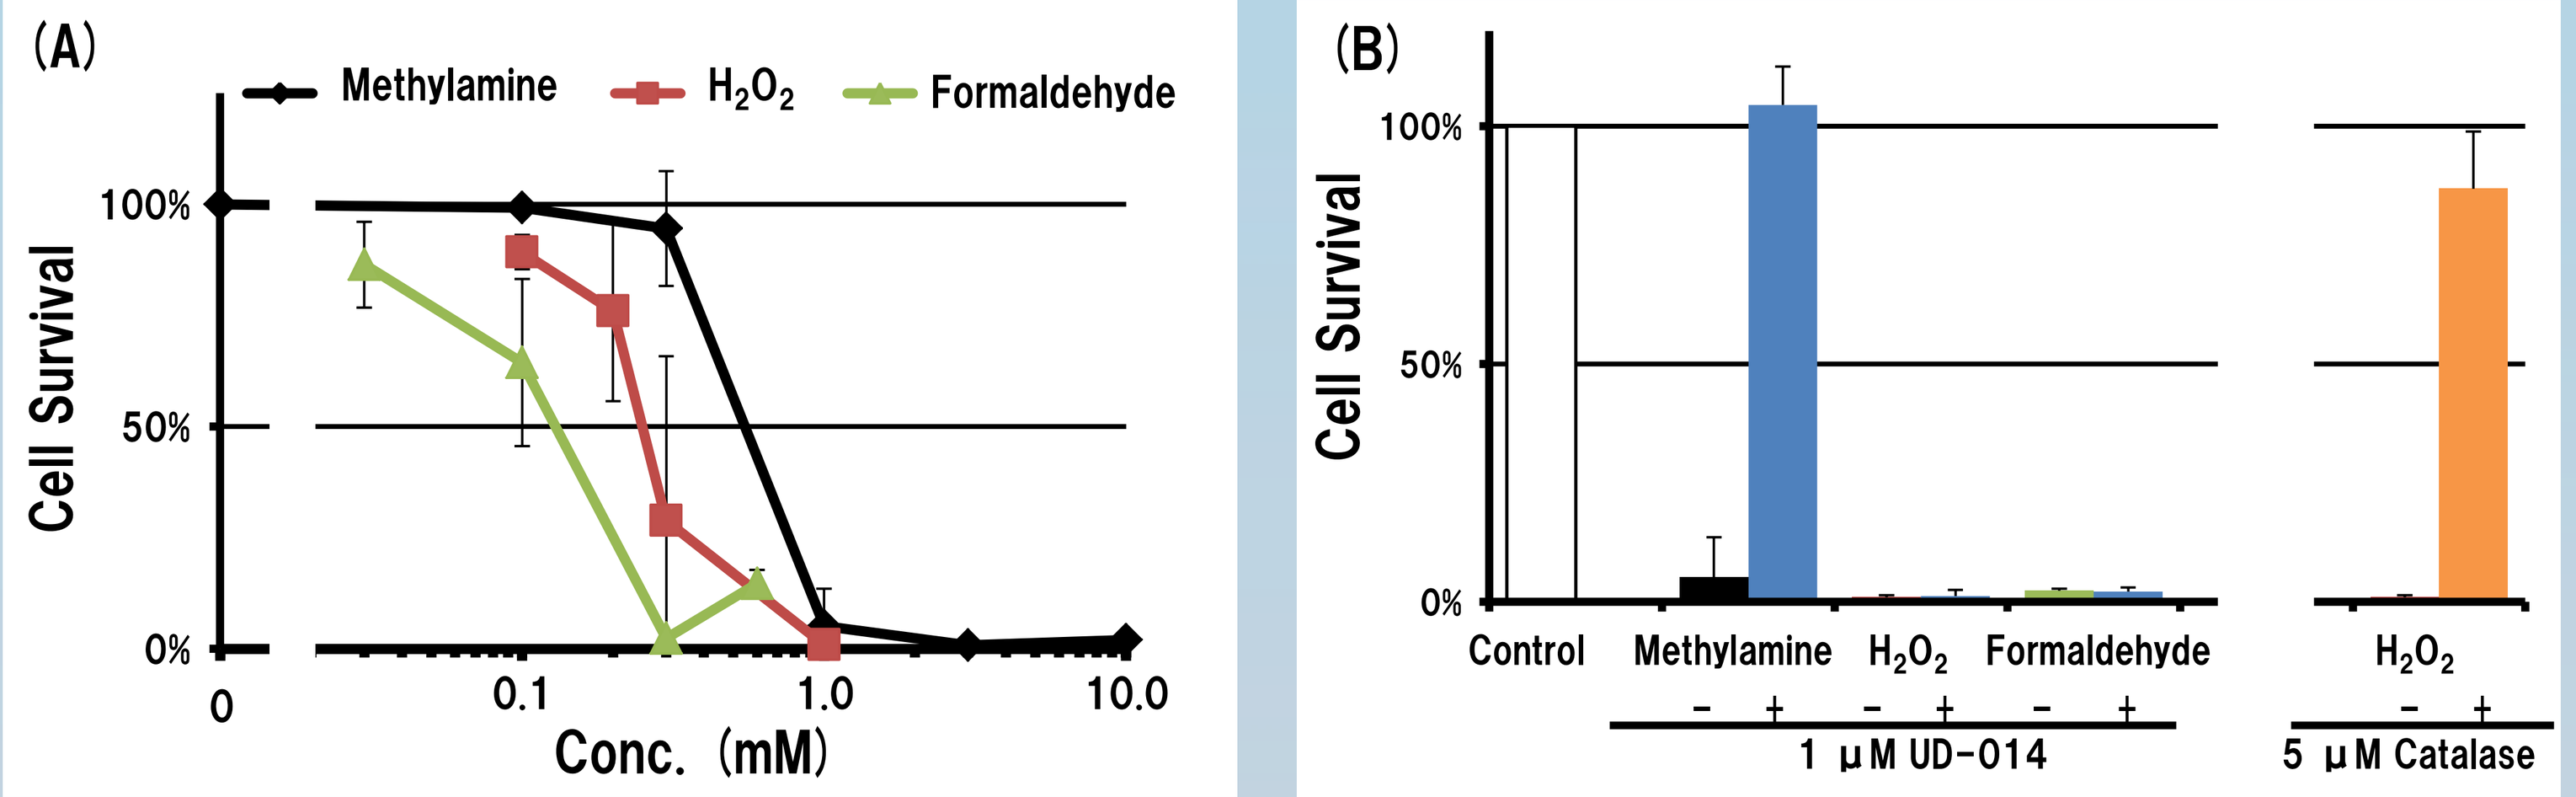
(A)
Methylamine
H2O2
Formaldehyde
Cell Survival
Conc. (mM)
(B)
Cell Survival
Control
Methylamine
H2O2
Formaldehyde
1 μM UD-014
5 μM Catalase

Fig.4 Methylamine metabolites induced endothelial cell toxicity in 2-day culture. (A)Metabolites induced cell toxicity at lower concentrations than methylamine. (B)UD-014 specifically rescued the methylamine-induced cellular toxicity. Methylamine:1 mM, H_2O_2:1 mM, Formaldehyde:0.3 mM.

SSAO Expression

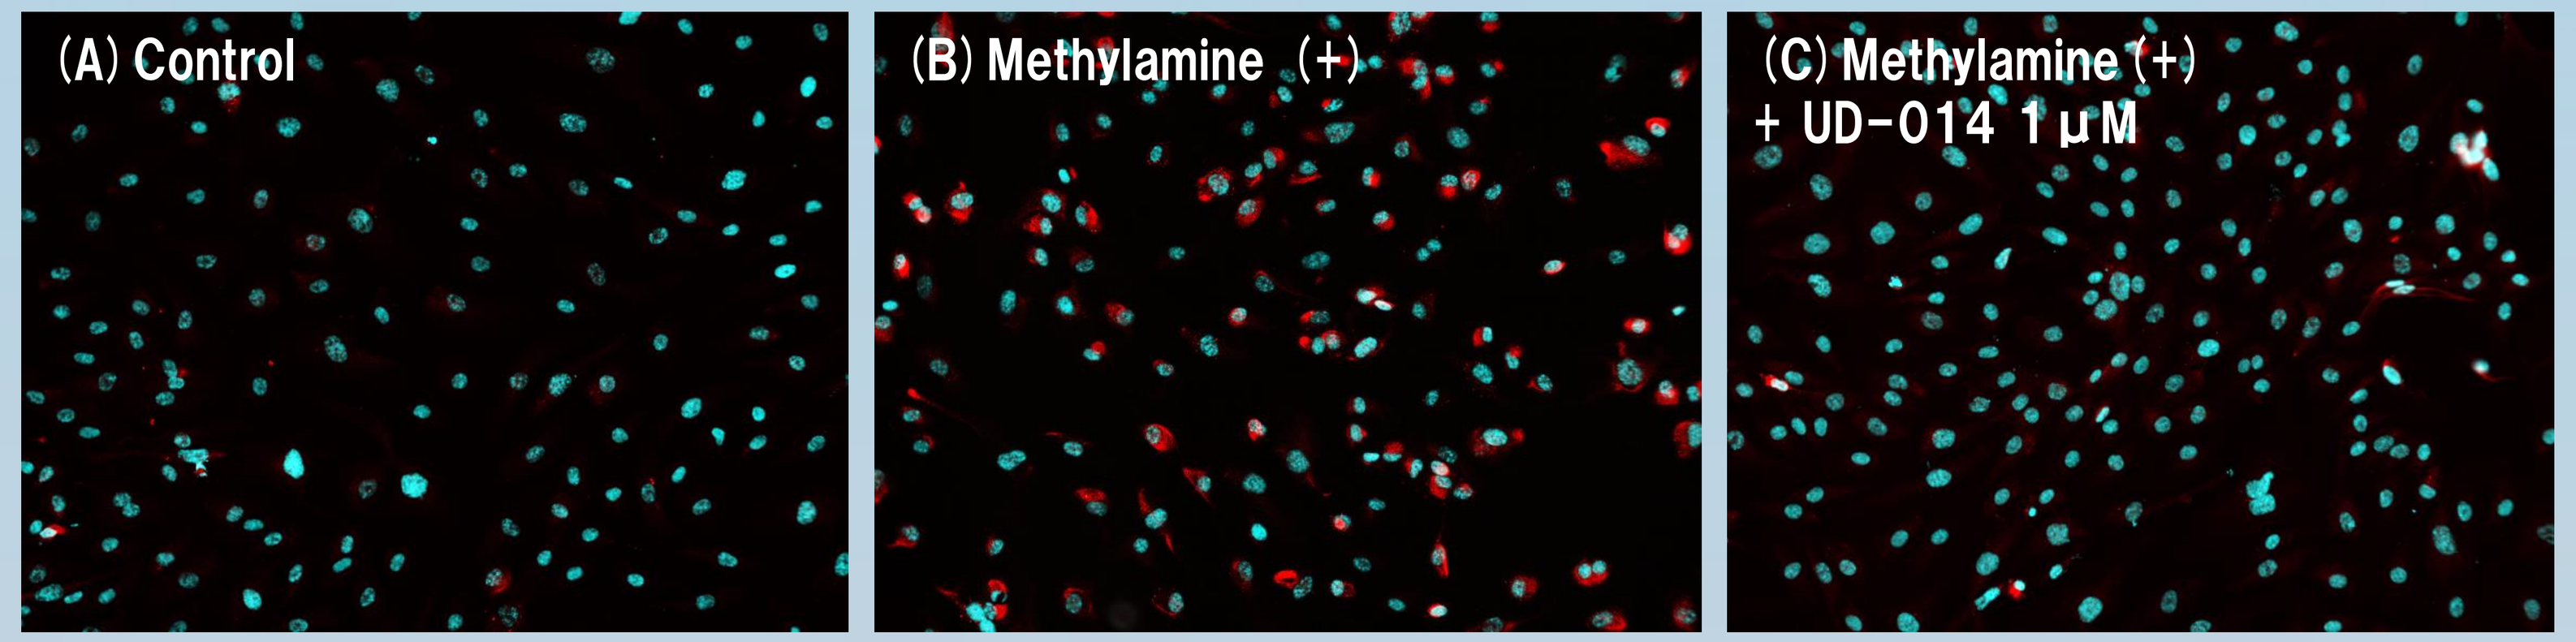
(A) Control
(B) Methylamine (+)
(C) Methylamine (+)
+ UD-014 1 μM

Fig.9 Methylamine-induced SSAO protein expression in GMVECs. Light blue : nucleus, red : SSAO protein.
(A) Vehicle control. (B) Methylamine treatment at 1 mM for 33 hr. Most cells expressed SSAO proteins. (C) 1 μM UD-014, treated before methylamine addition, inhibited SSAO protein expression efficiently.

Conclusion

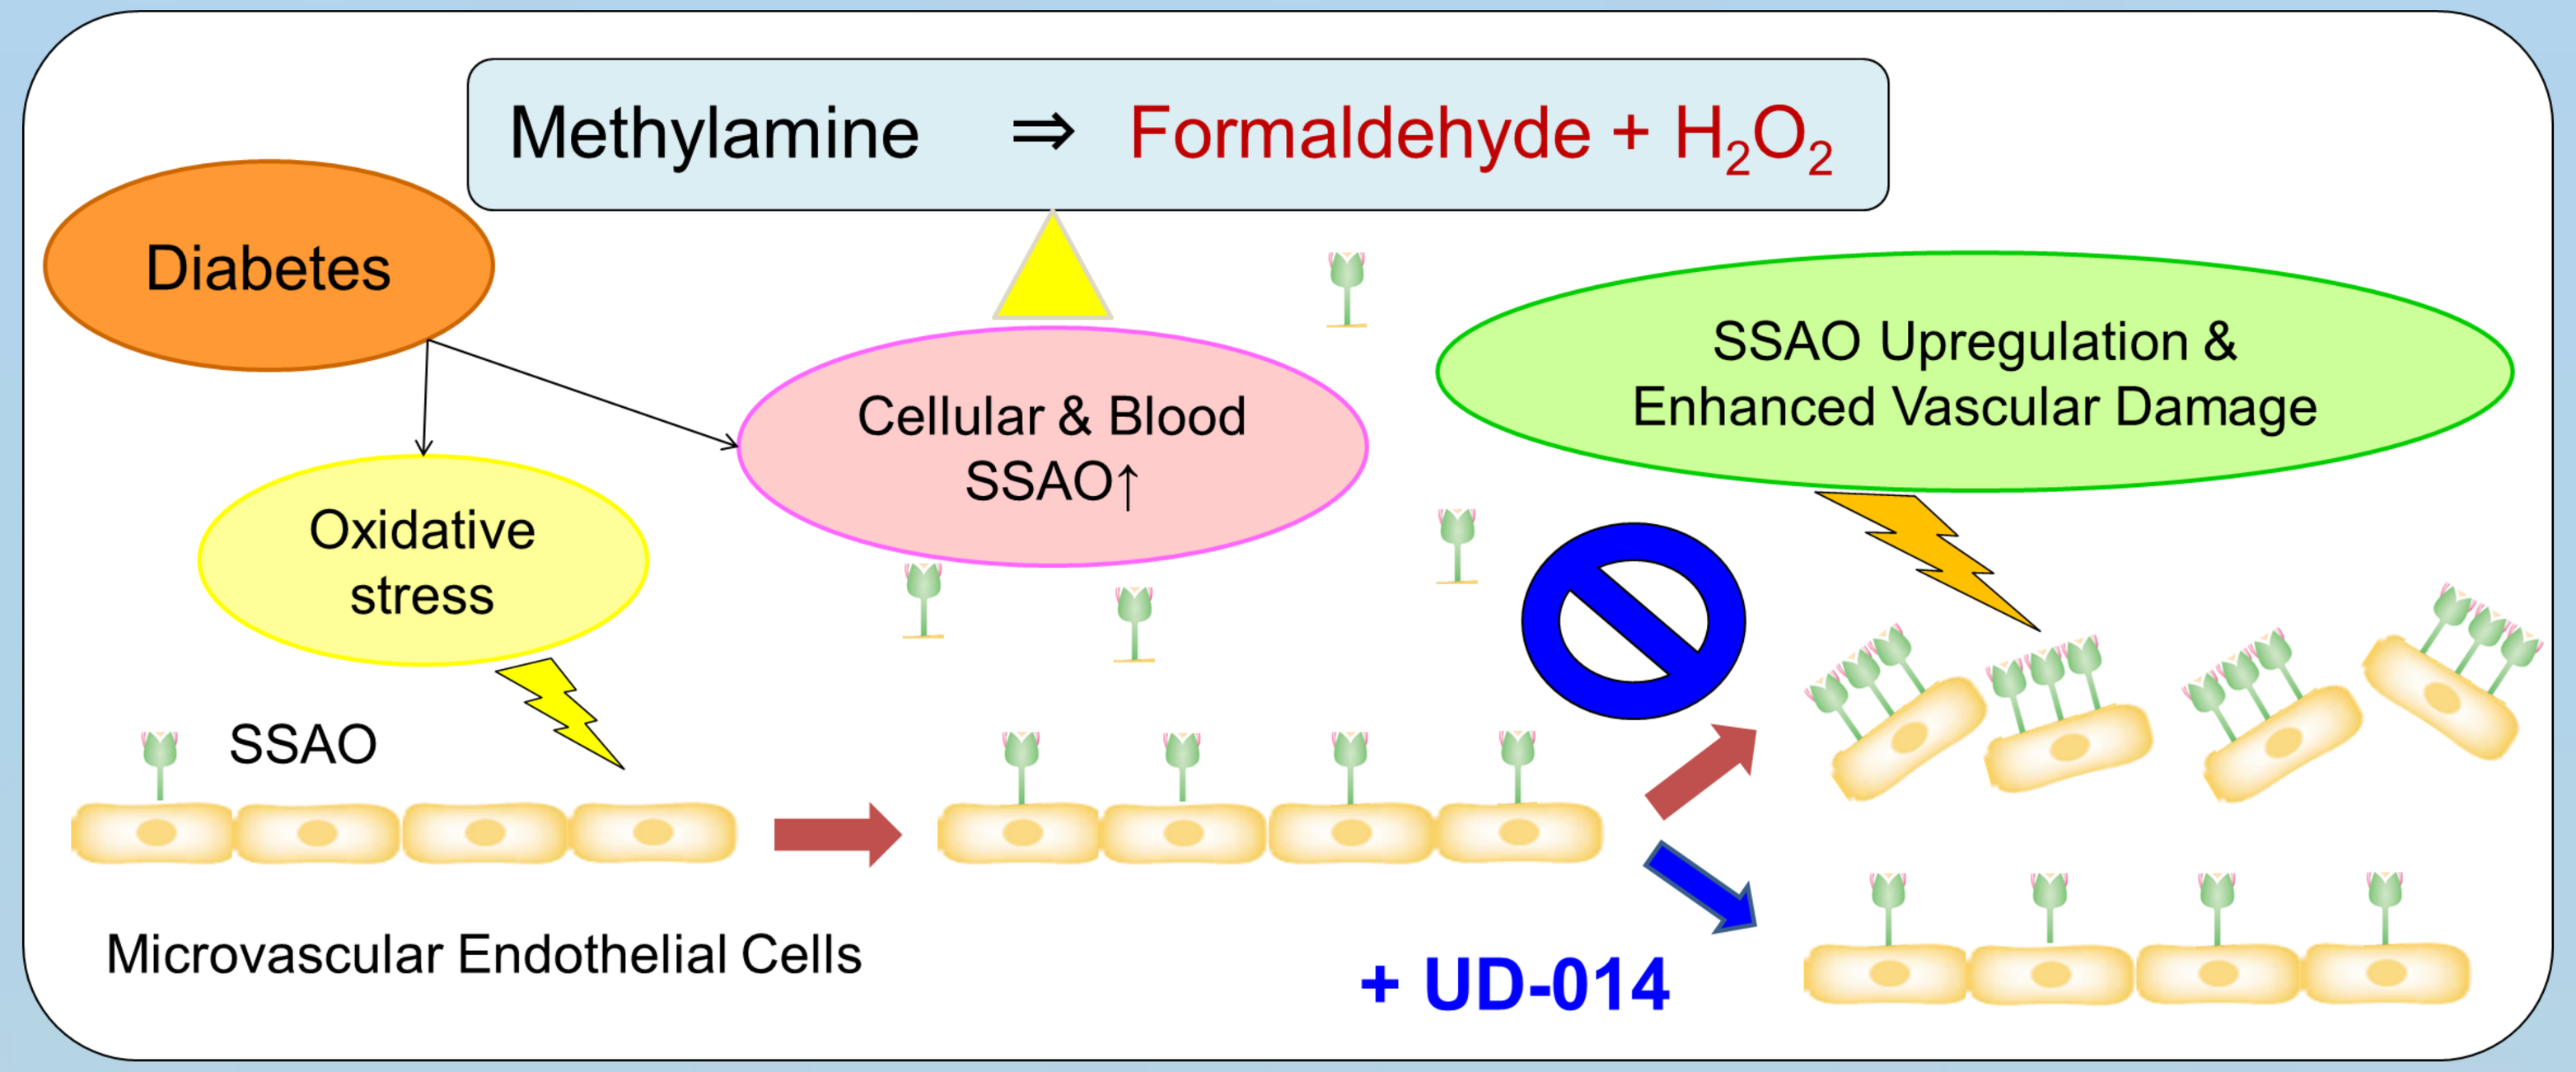
Methylamine ⇒ Formaldehyde + H2O2
Diabetes
Oxidative stress
Cellular & Blood SSAO↑
SSAO Upregulation & Enhanced Vascular Damage
SSAO
Microvascular Endothelial Cells
+ UD-014

SSAO upregulation may be a key to the damage of vascular endothelial cells in the presence of its substrates and contribute to the microvascular injury in diabetic complication. SSAO may be an in intriguing therapeutic target for diabetic nephropathy and chronic kidney diseases.

UD-014 may be a useful agent directly protecting the kidney from oxidative stress.

References

1. P.H. Yu, J. Neural Transm. Suppl., 52 (1998) 201– 216.
2. M. Salmi, S. Jalkanen, Nat. Rev. Immunol., 5 (2005) 760 –771
3. F. Yraola, et al., ChemMedChem 2(2007) 173 – 174.
4. H. Garpenstrand, et al., Diabet. Med. 16 (1999) 514– 521.
5. M.S. Lin, et al., Clinical Biochemistry 41 (2008) 1362–1367.
6. P.H. Yu, D.M. Zuo, Atherosclerosis 120 (1996) 189– 197.
7. P.H. Yu, D.M. Zuo, Diabetes. 42, (1993):594-603.

COI Disclosures, All authors: UBE Employees

Contact information: Name: Shigeru Ushiyama E mail: 34752u@ube-ind.co.jp

APPRISOR™